32nd IFSCC Congress London 19-22 September 2022

CRODAROM
Part of Croda International Plc

Natural solution for a skin microbiome friendly global care and odour reducer

597

Fischer Desnos Amandine[1], Gely Delphine[1], Chollet Dominique[1], Soléau Corinne[1]*
[1]Crodarom, Chanac, France

Introduction:

In this quest for mental and physical well-being, it is essential today for consumers to accept their appearance, their body by taking care of it as a whole: to be "Body positive". A new wave of holistic, honest, and inclusive products is coming to market to challenge the dictates of society. It's time to upgrade our daily body care routines and treat it with high-quality, high-performance, microbiome-friendly ingredients typically reserved for the face.
Human scent is genetically controlled and systemically influenced by gender, ethnicity, along with emotional, physiological, and environmental factors (influence of the composition and quantity of sweat). With ageing, the diversity of odour causing bacteria, *Corynebacterium* mainly, increases and body odour changes (1) (2). Body malodour, including foot odour, suppresses social interaction by diminishing self-confidence (3) (4).
Zanthoxylum bungeanum fruit extract (ZBFE) is evaluated for its global body care actions with multifunctional properties particularly for its malodour control and skin microbiota friendly capacity.

Materials & Methods:

SNIFF TEST. The underarm odour intensity is self-evaluated from T0 to 8 hours after application (T8) of a roll-on formulation containing 3.0% ZBFE versus placebo. Volunteers are asked not to eat spicy food, onions, or garlic, and not to use any perfumed products. Scores from 5 very good odour to 0 none are attributed every hour. 14 volunteers are recruited, with 2 evaluations per volunteer, which makes it to a total of 28 set of data.
INHIBITION OF LIPASE FROM *CORYNEBACTERIUM XEROSIS*. Liposoluble glyceryl tributyrate used as a substrate of lipase is hydrolysed in glycerol and hydrosoluble butyric acid. The trouble of the culture medium of *C.xerosis* disappears proportionally with the hydrolysis of the glyceryl tributyrate and therefore with an increased lipase activity. This trouble is measured at 620nm by spectrophotometry.
ASTRINGENCY CAPACITY. Based upon tannin-polymer interaction resulting in the formation of insoluble polymer tannin complexes which then precipitates. The difference in optical density (OD) before and after precipitation corresponds to the astringency capacity of the product. Samples are incubated with and without Methylcellulose and the OD at 280nm is measured by spectrophotometer.
SKIN MICROBIOTA FRIENDLY CAPACITY. The inhibitory capacity on skin microbiota of ZBFE is evaluated for 48H in the presence of *Staphylococcus epidermis* (ATCC 12228), and *Corynebacterium xerosis* (ATCC 373). The first colonises predominantly the axillae, head, and nares. It may have probiotic functions by preventing colonisation of more pathogenic bacteria. The second is mostly innocuous, commonly existing in commensal relationships with its hosts. It is very much implicated in armpit malodour.
HYDRATING EFFECT. - Human Epidermal Keratinocytes (HaCaT) are cultivated for 3 days with and without ZBFE. Hyaluronic acid synthesis is quantified by ELISA.
Normal Human Epidermal Keratinocytes (NHEK) differentiation at confluence is followed up visually by phenotype observation in the presence or absence of ZBFE.
ANTIOXIDANT CAPACITY. The oxydo-reduction decolourisation of DPPH to yellow DPPH-H and O_2 singlet scavenging capacity of ZBFE is followed by spectrophotometry.
SOOTHING EFFECT. Normal Human Dermal Fibroblasts (NHDF) are incubated with the test product for 24H. They are then irradiated or not with UVB at 60mJ/cm^2 and re-incubated with the product for another 24H. PGE_2 and IL6 synthesised are quantified by ELISA.

Results & Discussion:

SNIFF TEST.
Evaluation of the malodour neutralisation performance of ZBFE by determining the intensity of body odour on volunteers. (*p≤0.01)

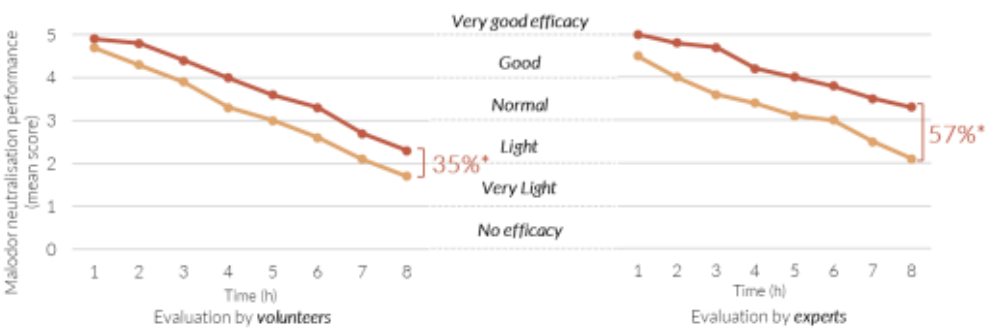
Negative Control
Roll-on 3% ZBFE
Malodor neutralisation performance (mean score)
Very good efficacy
Good
Normal
Light
Very Light
No efficacy
35%*
57%*
Time (h)
Evaluation by volunteers
Evaluation by experts

ZBFE's cream statistically significantly **decreases armpit malodour** after 8 hours by **35%***. Another trial has been carried out with 5 experts panellists, trained, and used to odour and perfumes evaluation. Expert evaluation of ZBFE's cream, shows a statistically significant **decrease of armpit malodour** after 8 hours by **57.1%***.

INHIBITION OF LIPASE FROM *CORYNEBACTERIUM XEROSIS*.

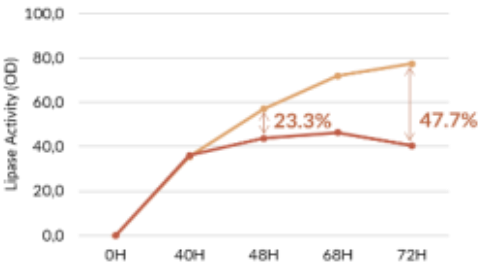
Lipase Activity (OD)
23.3%
47.7%
0H
40H
48H
68H
72H

Evaluation of the lipase inhibitory activity of 3% ZBFE on *Corynebacterium xerosis*.
Negative Control
ZBFE
ZBFE at 3% statistically significantly inhibits *C.xerosis* lipase by **47.7%*** at 72H (**p*<0.0001).

ASTRINGENCY CAPACITY.

	Mean MCP Tannin (% eq epicatechin)	Standard Deviation	% Variation vs Tannic wine
Tannic Wine (n=12)	1 334	222	Reference
ZBFE (n=4)	5 276	161	295%*

ZBFE shows a statistically higher astringent capacity of **295%*** than tannic red wine in the experimental condition of this assay (**p*<0.0001).

SKIN MICROBIOTA FRIENDLY CAPACITY.
Corynebacterium xerosis:.. <14%
Staphylococcus epidermis:.. <20%
ZBFE shows no antimicrobial properties on *C.xerosis* nor on *S.epidermidis* at the tested concentration.

HYDRATING EFFECT.

Product	Conc	Variation (%)	Mean HA Variation (%)
CTRL	-	Ref	Ref
ZBFE (n=5)	0.14%	45%* 52%*	49%
	0.26%	159%* 257%*	208%

ZBFE increases statistically significantly by **+208%*** Hyaluronic acid synthesis at 0,26% in the experimental condition of this assay (**p*<0.01).
It also increases keratinocytes **differentiation and keratinisation** (data not shown here).

ANTIOXIDANT CAPACITY.

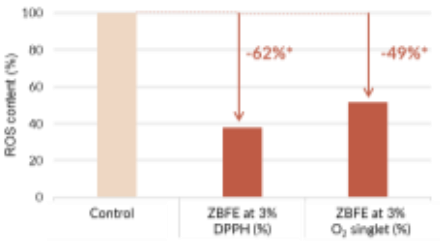
ROS content (%)
-62%*
-49%*
Control
ZBFE at 3% DPPH (%)
ZBFE at 3% O2 singlet (%)

ZBFE decreases statistically significantly by **62%*** DPPH and **49%*** O_2 singlet in the experimental condition of this assay (**p*<0.01).

SOOTHING EFFECT.

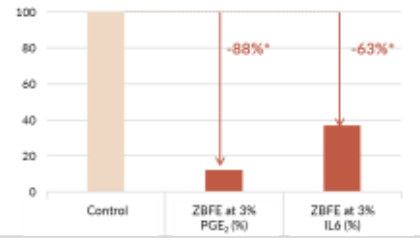
-88%*
-63%*
Control
ZBFE at 3% PGE2 (%)
ZBFE at 3% IL6 (%)

ZBFE decreases statistically significantly by **88%*** PGE_2 and **63%*** IL6 with UV induction in the experimental condition of this assay (**p*<0.01).

Conclusions:

It is now generally accepted that skin bacteria cause body odour by biotransformation of sweat components secreted in the human axillae. This **biotransformation** is generated by enzymes like lipases secreted by bacteria for their metabolism creating among others VFA (volatile fatty acids) (5) (6). Currently it is believed that benefits of healthy skin microflora are comparable with those of healthy gut flora. Low diversity has also been associated with some dysbiotic skin disorders (7) (8). Therefore, **preserving it**, seems increasingly important. There is therefore a significant shift from the current reliance of deodorants on fragrances and broad-spectrum antimicrobial agents. A new generation of deodorant systems appears based on targeting specific bacteria, metabolic pathways or key enzymes. *Zanthoxylum bungeanum* fruit extract (ZBFE) answers to this demand being a full body care product with a microbiota friendly action through inhibition of bacterial enzyme that generate odoriferous compounds and with astringent, protective, hydrating, and soothing properties.

Acknowledgements:

The authors acknowledge Michael White and Manuel Serna from Iberchem SA based in Alcantarilla for their precious help for the sniff test.

References:

1. Li M, Budding AE, van der Lugt-Degen M, Du-Thumm L, Vandeven M, Fan A. The influence of age, gender and race/ethnicity on the composition of the human axillary microbiome. International Journal of Cosmetic Science. 2019, Vol. 41, pp. 371–377.
2. Mitro S, Gordon AR, Olsson MJ, Lundström JN. The Smell of Age: Perception and Discrimination of Body Odors of Different Ages. PLoS ONE. 2012, Vol. 7, 5, p. e38110.
3. Troccaz, MC. The biosynthetic pathway of sulfur-containing molecules in Human Axillary Malodor: from precursors to odorous volatiles. Univ. Genève : Thèse de doctorat no. Sc. 4102, 2009.
4. Martin A, Hellhammer J, Hero T, Max H, Schult J, Terstegen L. Effective prevention of stress-induced sweating and axillary malodour formation in teenagers. International Journal of Cosmetic Science. 2011, Vols. Effective prevention of stress-induced sweating and axillary malodour formation in teenagers Martin A, Hellhammer J, Hero T, Max H, Schult J, Terstegen L. International Journal of Cosmetic Science, 2011, 33, 90–97, pp. 90–97.
5. James AG, Austin CJ,Cox DS, Taylor D and Calvert R. Microbiological and biochemical origins of human axillary Odour. FEMS Microbiol Ecol. 2013, Vol. 83, pp. 527–540.
6. Barzantny H, Brune I, Tauc A. Molecular basis of human body odour formation: insights deduced from corynebacterial genome sequences. International Journal of Cosmetic Science. 2012, Vol. 34, pp. 2-11.
7. Grice, EA. The skin microbiome: potential for novel diagnostic and therapeutic approaches to cutaneous disease. Semin Cutan Med Surg. 2014 Jun, Vol. 33, 2, pp. 98–103.
8. Scharschmidt, TC. Establishing Tolerance to Commensal Skin Bacteria. Dermatol Clin. 2017 Jan, Vol. 35, 1.

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET