"WHERE BEAUTY, SCIENCE AND INNOVATION MEET"
32nd IFSCC Congress
London 19–22 September 2022

DUCRAY LABORATOIRES DERMATOLOGIQUES

Pierre Fabre Dermo-Cosmétique & Personal Care Recherche & Développement

Silybum marianum extract, Manganese PCA and *Lespedeza capitata* extract are active on hair growth and anchorage in human hair follicle dermal papilla cells

Poster 395

D. Bacqueville[1], C. Mas[1], M. Lévêque[1], M.J. Haure[1], A. Noustens[1], V. Mengeaud[2], S. Carrère[1], S. Bessou-Touya[1] and H. Duplan[1]

[1] R&D department, Pierre Fabre Dermo-Cosmétique, Toulouse, France - [2] Laboratoire dermatologique Ducray, Lavaur, France

Introduction:

Hair loss impacts significantly the quality of life of patients and is one of the most common cause of dermatological consultations. In recent years, progress has been made to diagnose but also treat hair defects. However, it remains to better understand the pathophysiology of hair loss for improving treatments (1).

The hair follicle (HF) undergoes cycles of growth (anagen), regression (catagen) and rest (telogen) phases. Both chronic and reactive hair loss are linked to a dysfunction of the HF cycle leading to a premature hair loss (2, 3). Therefore, the use of specific active ingredients targeting the HF could improve hair loss (4, 5).
Hair growth cycle is a highly regulated process during which different compartments of HF maintain close relations through many molecular communications. Among these exchanges, signals emitted by the dermal papilla play a central role (6).

Therefore, the aim of this study was to evaluate the efficacy of three active ingredients on hair growth and anchorage in dermal papilla cells isolated from human hair follicle (HFDPC). The first active ingredient was a new patented extract from *Silybum marianum* containing less than 2% silymarin (SME) (*WO/2021/023820*). It has been shown to stimulate the expression of the keratin 75 (K75), a specific marker of the "companion layer" on the outer root sheath of the HF which is involved in the anchorage of the hair shaft (7). K75 deficit is also associated with a rare genetic alopecia: loose anagen hair syndrome. Manganese PCA (MnPCA) and a *Lespedeza capitata* extract (LCE) were also tested. LCE was also recently patented (*WO/2020/020791A1*).

Materials & Methods:

Human dermal papilla cell culture and active ingredients

All experiments were performed by using Human Follicle Dermal Papilla Cells (HFDPC) isolated from different donors. Cells were seeded in an adapted culture medium with specific complements as recommended by the supplier. HFDPC were grown 24h to 48h to reach about 80% confluency before active ingredients treatment. Active ingredients were obtained from Pierre Fabre Laboratories after specific extraction.

Growth factor receptor signaling pathways evaluation

The growth factor receptor (GFR) signaling pathways were analysed by using Proteome profiler™ antibody arrays that allow the specific detection of tyrosine phosphorylation of human receptor tyrosine kinases and their downstream protein kinases. HFDPC cells were stimulated for 1h with SME before harvesting and cell lysate analysis.

Wnt/β catenin signaling pathway evaluation

Wnt/β catenin pathway was studied with a gene reporter assay. HFDPC were transfected with a lentivirus expressing a luciferase gene under the control of Wnt/β catenin promoter (TCF/LEF transcriptional response element). Cells were incubated 24h with MnPCA and luminescence was quantified by using Bright Glo™ substrate.

Versican, VEGF and DKK1 release evaluation

ELISA tests were performed to quantify the production of Versican, VEGF and DKK1 from the HFDPC culture medium. Versican and VEGF release was analyzed following MnPCA treatment during 48h and 24h, respectively. The production of DKK1 was also measured 24h after HFDPC incubation with LCE.

5α reductase activity evaluation

The 5α reductase (5αR) enzyme metabolizes testosterone into dihydrotestosterone (DHT) that activates androgen receptor, especially in androgenetic alopecia. HFDPC were treated with LCE extract for 24h. Then, 5αR activity was evaluated by performing testosterone metabolism studies with [^{14}C]-testosterone and metabolites quantification on thin layer chromatography.

Data analysis and Statistics

Data were obtained from 2-4 donors and performed in duplicate or triplicate according to the different readouts. Results are presented as mean ± standard deviation. The inter-group comparison was performed by a One-way Anova completed by a Dunnett's multiple comparison test on normalized data comparatively to the untreated control. Statistical analysis was performed with PRISM software: * $p<0.05$ and ** $p<0.01$.

Results:

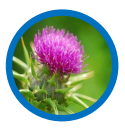

Silybum marianum extract

Growth stimulation
Receptor signaling

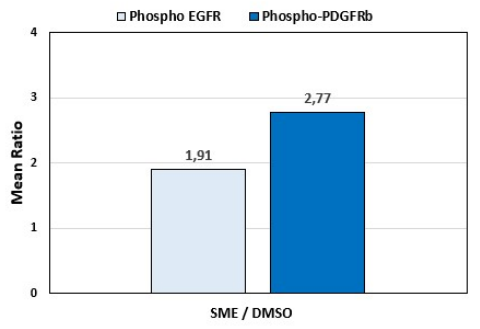
Phospho EGFR
Phospho-PDGFRb
Mean Ratio
1,91
2,77
SME / DMSO

Growth stimulation
Downstream signaling

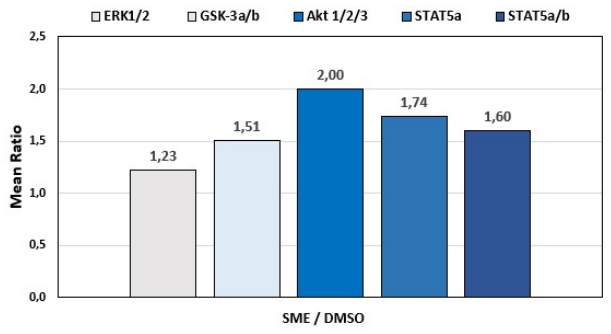
ERK1/2
GSK-3a/b
Akt 1/2/3
STAT5a
STAT5a/b
Mean Ratio
1,23
1,51
2,00
1,74
1,60
SME / DMSO

SME 30 µg/mL showed activation of GFR signaling pathways in HFDPC. The tyrosine phosphorylation of the EGFR and PDGFR were specifically and respectively induced by 1.9 and 2.8-fold following 1h stimulation in the presence of SME. Moreover, the downstream effectors of these GFR were also increased and they were upregulated from 1.2 to 2-fold for ERK1/2, GSK3 α/β, Akt1/2/3 and STAT5 α/β after SME treatment.

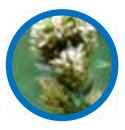

Lespedeza capitata extract

Growth extension
DKK1 production

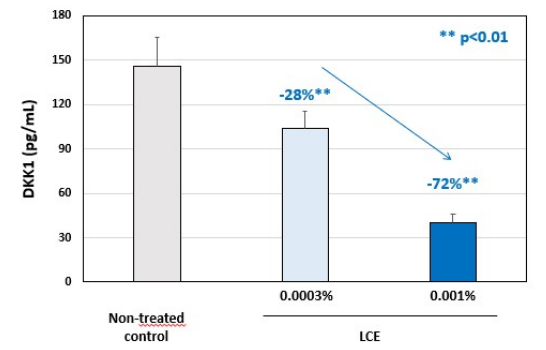
DKK1 (pg/mL)
** p<0.01
-28%**
-72%**
Non-treated control
0.0003%
0.001%
LCE

Anti-androgenic effect
5αReductase activity

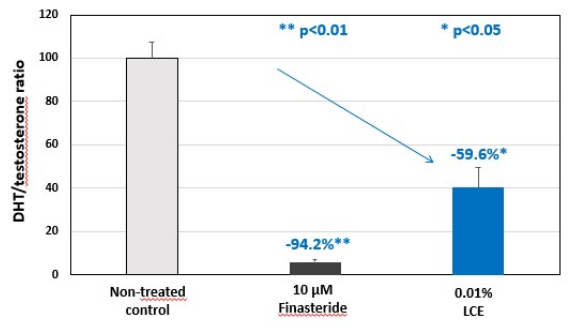
DHT/testosterone ratio
** p<0.01
* p<0.05
-94.2%**
-59.6%*
Non-treated control
10 µM Finasteride
0.01% LCE

LCE reduced the release of DKK1, a Wnt signaling inhibitor. The inhibition reached 72% at 0.001%. Finally, this active ingredient was also able to decrease the 5αR activity with a 59.6% inhibition at 0.01% dose.

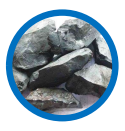

Manganese PCA

Growth extension
Wnt/β catenin activation

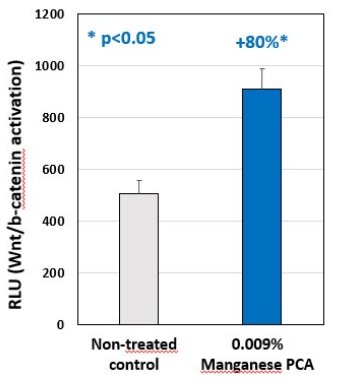
RLU (Wnt/b-catenin activation)
* p<0.05
+80%*
Non-treated control
0.009% Manganese PCA

MnPCA stimulated the Wnt/β catenin pathway up to 80% at 0.009%. It also boosted the release of both Versican and VEGF in the HFDPC culture medium. The production of the matricial proteoglycan Versican was induced by 33-fold at a 0.01% dose whereas the angiogenic factor VEGF was stimulated up to 3.3-fold at 0.009%.

Anchorage
Versican production

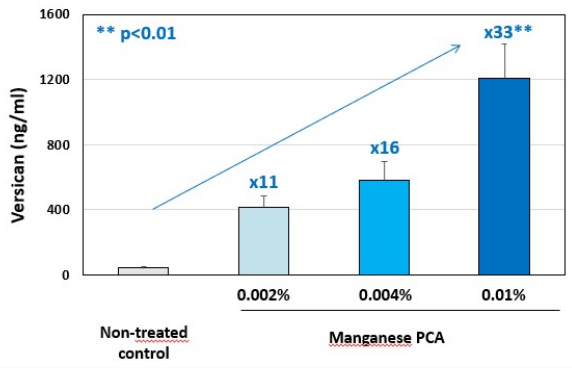
Versican (ng/ml)
** p<0.01
x11
x16
x33**
Non-treated control
0.002%
0.004%
0.01%
Manganese PCA

Microcirculation stimulation
VEGF production

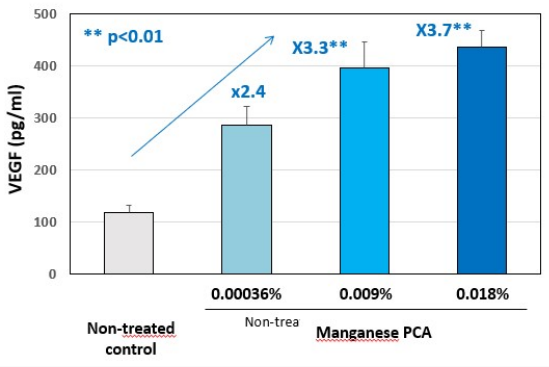
VEGF (pg/ml)
** p<0.01
x2.4
X3.3**
X3.7**
Non-treated control
0.00036%
0.009%
0.018%
Manganese PCA

References:

1. Jeffrey E Plowman, Duane P Harland. The Follicle Cycle in Brief. Review Adv Exp Med Biol. 2018;1054: 15-17.
2. Gentile P, Garcovich S. Advances in Regenerative Stem Cell Therapy in Androgenic Alopecia and Hair Loss: Wnt pathway, Growth-Factor, and Mesenchymal Stem Cell Signaling Impact Analysis on Cell Growth and Hair Follicle Development. Cells. 2019 May 16;8(5): 466.
3. Choi BY. Targeting Wnt/β Catenin Pathway for Developing Therapies for Hair Loss. Int J Mol Sci. 2020 Jul 12;21(14): E4915.
4. Jungyoon Ohn, Kyu Han Kim, Ohsang Kwon. Evaluating Hair Growth Promoting Effects of Candidate Substance: A Review of Research Methods. Review J Dermatol Sci. 2019 Mar;93(3): 144-149.
5. Anna Herman, Andrzej P Herman. Topically Used Herbal Products for the Treatment of Hair Loss: Preclinical and Clinical Studies. Review Arch Dermatol Res. 2017 Oct;309(8): 595-610.
6. Madaan A, Verma R, Singh AT, Jaggi M. Review of Hair Follicle Dermal Papilla cells as in vitro screening model for hair growth. Int J Cosmet Sci. 2018 Oct;40(5): 429-450.
7. Mesler AL, Veniaminova NA, Lull MV, Wong SY. Hair Follicle Terminal Differentiation Is Orchestrated by Distinct Early and Late Matrix Progenitors. Cell Rep. 2017 Apr 25;19(4):809-821.

Conclusions:

In this study, the efficacy of three active ingredients was evaluated on hair growth and anchorage by using HFDPC as a pre-clinical model. Results have shown that SME modulates the cell growth by acting on EGFR/PDGFR signaling pathways. In addition, MnPCA and LCE enhanced the anagen phase via the Wnt/β catenin pathway. MnPCA also improved HF anchorage and microcirculation by stimulating Versican and VEGF production, respectively. Finally, LCE favored DHT decrease and inhibited androgen metabolism. Altogether, the data suggest that a combination of SME + MnPCA + LCE may be useful to improve hair loss treatment by a specific action on both hair growth and anchorage.

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET