32nd IFSCC Congress
London 19–22 September 2022
"WHERE BEAUTY, SCIENCE AND INNOVATION MEET"

Mantecorp skincare

CIDP NATURALLY INNOVATIVE

Randomized study to assess the efficacy of a facial cosmetic product with nanoencapsulated cysteamine in women presenting melasma

Poster 472

VALPAÇOS Camila[1], LOPES, Luana[1], SVOBODA Diego[1] , MIRANDA, Daniele[1], FERNANDES Erika[1], PRESTI Carla[2] , AYRES, Eloisa[3]

1 Centre International de Développement Pharmaceutique, Rio de Janeiro, Brazil
2 Mantecorp Skincare, São Paulo, Brazil
3 DermScience Study Center, Rio de Janeiro, Brazil

Introduction:

Melasma is a common and persistent pigmentary disease that particularly affects women during their reproductive period [1-2]. The chronicity of the disease and the involvement of visible areas during working age generates a great negative impact on the quality of life of affected individuals.[3]

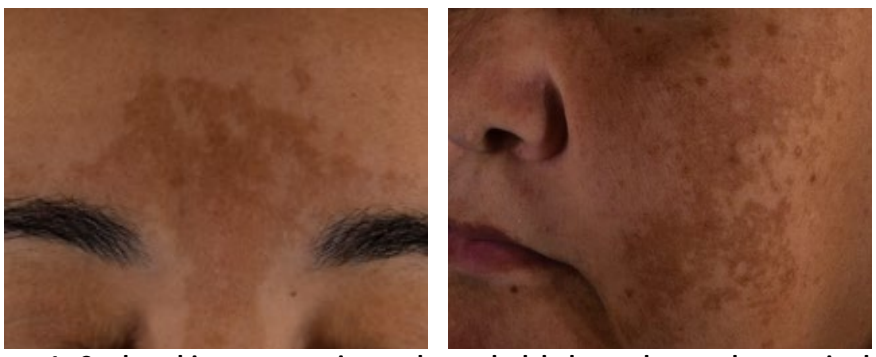

Figure 1- Study subjects presenting malar and glabelar melasma characterized by symmetrical hyperpigmented macules or patches with irregular borders, most often distributed on the face.

There are several therapeutic options for the treatment of melasma that act on different stages of melanogenesis. The vast majority of effective depigmenting agents are tyrosinase inhibitors, of which hydroquinone is the most studied and most effective drug. However, there is concern about its tolerability and its prolonged use due to the risk of adverse events such as exogenous ochronosis. Therefore, there is an interest in seeking other depigmenting agents in the treatment of melasma. [4-5]

Materials & Methods:

This single-blind, randomized and comparative study of a cosmetic product with nano encapsulated cysteamine, a promising and more tolerable option, and sunscreen SPF 60 versus sunscreen SPF 60 alone was conducted to investigate the efficacy in improving melasma and improving the quality of life of subjects.

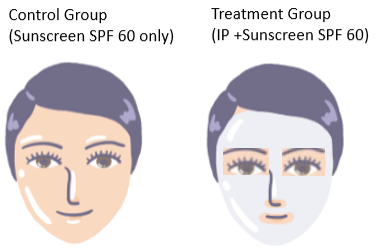
Control Group
(Sunscreen SPF 60 only)
Treatment Group
(IP +Sunscreen SPF 60)

Figure 2- Control group was oriented to apply sunscreen SPF 60 twice a day on entire face for 84 days. Treatment group was oriented to apply the IP on entire face every night and apply sunscreen SPF 60 twice a day for 84 days.

Table 1- Subject's characteristics panel

	Investigational group (IP + sunscreen SPF 60)		Control group (sunscreen SPF 60)	
Mean age (years)	**45**		**44**	
Female	24	100,0%	19	100,0%
Phototype III (Fitzpatrick)	6	25,0%	3	15,8%
Phototype IV(Fitzpatrick)	13	54,2%	8	42,1%
Phototype V (Fitzpatrick)	5	20,8%	8	42,1%
Presenting Melasma	24	100,0%	19	100,0%
Chronic non-smokers	24	100,0%	19	100,0%

Table 2- Study flowchart representing the kinetics and study procedures

Study procedures	D0	D28	D56	D84
Dermatological evaluation (melasma hyperpigmentation intensity, area affected by melasma, skin tone uniformity, skin hydration, softness, brightness and oiliness).	X		X	X
MelasQoL questionnaire (impact of melasma on their quality of life)	X	X	X	X
Color Face standardized clinical pictures	X			X
Confocal Reflecting Microscopy (characteristics of the epidermis, dermo-epidermal junction, and dermis)	X			X
Possible local intolerance and adverse event evaluation		X	X	X

References:

1. Gupta A, Gover M, Nouri K, Taylor S. The treatment of melasma: A review of clinical trials. J Am Acad Dermatol. 2006;55(6):1048-65.
2. Rendon M, Berneburg M, Arellano I, Picardo M. Treatment of melasma. J Am Acad Dermatol. 2006;54(5):S272-81.
3. Pollo CF, Miot LDB, Meneguin S, et al. Factors associated with quality of life in facial melasma: a cross-sectional study. Int J Cosmet Sci 2018; 40: 313–316.
4. Tse TW. Hydroquinone for skin lightening: safety profile, duration of use and when should we stop? J Dermatolog Treat 2010; 21: 272–275.
5. Nordlund JJ, Grimes PE, Ortonne JP. The safety of hydroquinone. J Eur Acad Dermatol Venereol 2006; 20: 781–787.

Acknowledgements:

We would like to acknowledge the Mantecorp Skincare group for the partnership and clinical team at CIDP Brazil for contributing to the study conduction and report writing.

Results & Discussion:

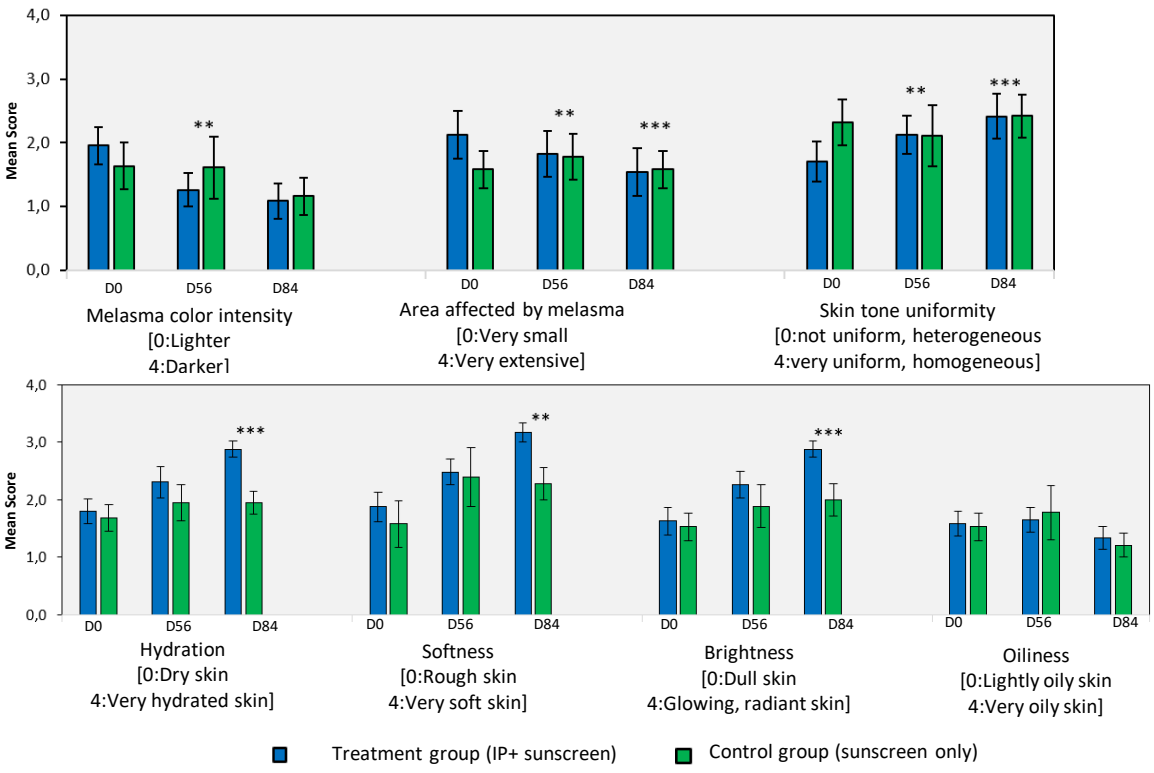
Mean Score
4,0
3,0
2,0
1,0
0,0
D0 D56 D84
Melasma color intensity
[0:Lighter
4:Darker]
Area affected by melasma
[0:Very small
4:Very extensive]
Skin tone uniformity
[0:not uniform, heterogeneous
4:very uniform, homogeneous]
Hydration
[0:Dry skin
4:Very hydrated skin]
Softness
[0:Rough skin
4:Very soft skin]
Brightness
[0:Dull skin
4:Glowing, radiant skin]
Oiliness
[0:Lightly oily skin
4:Very oily skin]
Treatment group (IP+ sunscreen)
Control group (sunscreen only)

Figure 3- Dermatological evaluation of skin parameters by group, Error bars: 95% CI, **p value≤0.010, *p value≤0.001**

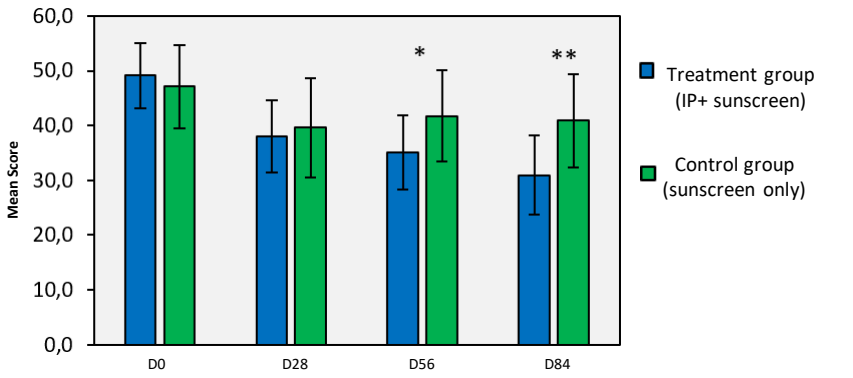
Mean Score
60,0
50,0
40,0
30,0
20,0
10,0
0,0
D0
D28
D56
D84
Treatment group (IP+ sunscreen)
Control group (sunscreen only)

Figure 4- MelasQoL questionnaire by group, Error bars: 95% CI, *p value≤0.050, **p value≤0.010

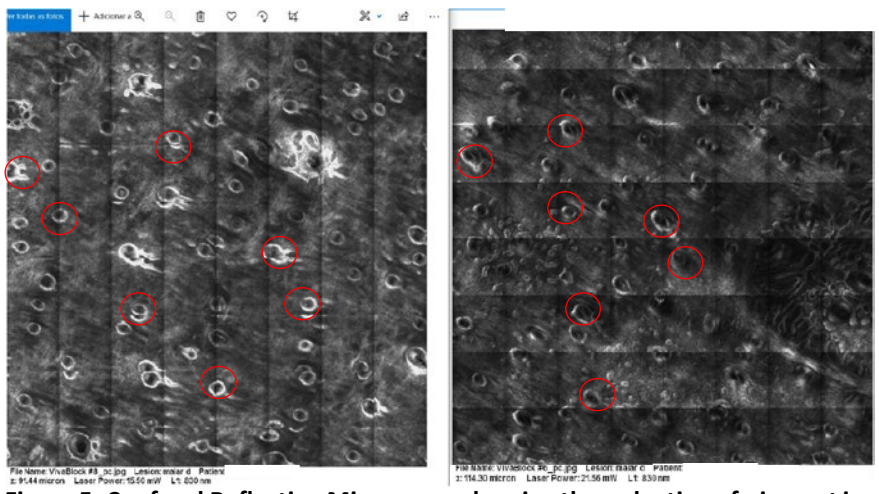

Figure 5- Confocal Reflecting Microscopy showing the reduction of pigment in skin keratinocytes. Subject allocated in treatment group. (Left- D0 and Right- D84) (red circles marking the reduction of white reflection that represents pigment)

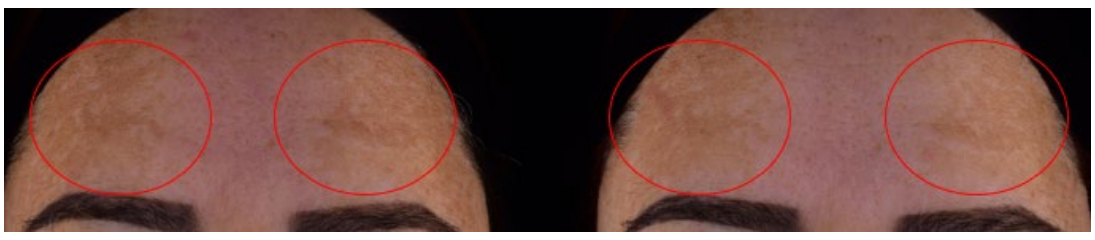

Figure 6- Color Face standardized clinical pictures. Subject allocated in treatment group. (Left- D0 and Right- D84) (red circles representing the hyperpigmentation improvement)

During the study, it was possible to observe on the comparison between two groups, a significant improvement of melasma hyperpigmentation intensity after 56 days (p value = 0.004) using investigational product plus sunscreen SPF 60 and a significant improvement of the area affected by melasma (p value <0.001), skin tone uniformity (p value = 0.001), skin hydration (p value <0.001), softness (p value =0.006) and brightness (p value <0.001) after 84 days using investigational product plus sunscreen SPF 60 when compared to the group sunscreen SPF 60 only **(Figure 3)**.
MelasQol results revealed a significant difference (p value = 0.003) in the impact of melasma on quality of life after 84 days after IP plus sunscreen SPF 60 use when compared to the group sunscreen SPF 60 only **(Figure 4)**.
Reflective confocal microscopy results showed pigment reduction in keratinocytes in the epidermis, around adnexal structures and at the dermal-epidermal junction **(Figure 5)**. ColorFace standardized pictures contributed with illustrative images of hyperpigmentation improvement **(Figure 6)**
No subject presented any medically relevant local intolerance that occurred at the assessment site (face) due product application.

Conclusions:

Nano encapsulated cysteamine improves hyperpigmentation intensity, area affected by melasma and individual's quality of life when compared to the control group, with a good tolerability profile, and is an efficient alternative to depigmenting agents such as tyrosinase inhibitors.

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET