LINKING SKIN BARRIER IMPROVEMENT TO UNDERLYING MOLECULAR MECHANISMS USING A MULTIOMICS APPROACH.

Poster 260

Arbey Eric[a], Fabre Aurélie[a] Baltenneck Carine[a], Rayée Chrystelle[a], Lynch Barbara[a], Sahuc Florent[b], Benas Damien[b], Valentin Tanguy[b], Ovigne Jean-Marc[b], Gregoire Sebastien[a], Potter Anne[a], Billoni Nelly[a], Bernard Dominique[a], Foucher Aude[a]*

a. *L'Oréal Research and Innovation, Aulnay-sous-Bois, France*

b. *Episkin, Lyon, France*

1 INTRODUCTION

The skin, the human body's largest organ, provides a protective barrier against multiple threats mainly penetration of molecules, water loss and exposome (i.e: UV, ozone). Reconstructed human epidermal (RHE) models have been developed to reproduce *in vitro* skin functions for research and evaluation purposes. SkinEthic™ and EPISKIN™ RHE models were commercialized in the early 1990s and validated for use in skin irritation and corrosion assays [1, 2]. Despite many advances in skin engineering and skin model reconstruction, there are still challenges remaining for the production of models with all *in-vivo* functions. One of the essential skin's functions which require further improvements in reconstructed models is the barrier function [3]. The barrier function *in-vivo* provides a- protection against the external environment (OUT-IN barrier, against physical, chemical and biological aggressors) and b- retention of moisture (IN-OUT barrier). The *in-vivo* barrier function is supported mainly by the *stratum corneum* (SC), a multilayer tissue composed of flattened anucleate corneocytes, surrounded by multiple planar lamellae layers, enriched in ceramides, cholesterol and free fatty acids (FFA) [4]. In this study, we evaluated the effect of adding a mix of lipids to the culture media of reconstructed skins on the physical aspects of barrier function, both OUT-IN and IN-OUT, as well as the mechanical properties of the models. We then determined the molecular entities that were associated with the improved function.

2

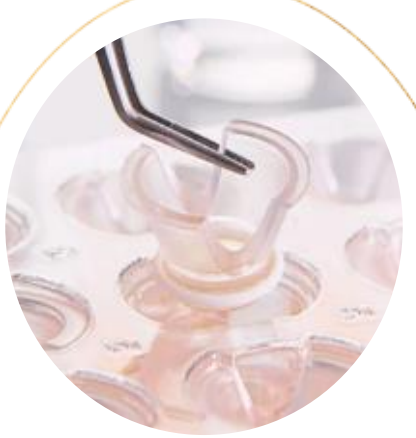

SkinEthic™ Reconstructed skin model
+/- FBA
D17

- **Histological analyses**

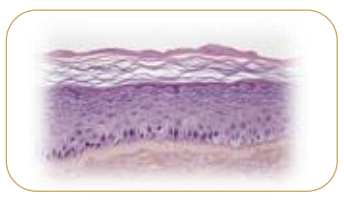

- **Barrier function mechanical and physical measurements**

Bodipy, Kp, TEWL, TEER, OCT and mechanical properties.

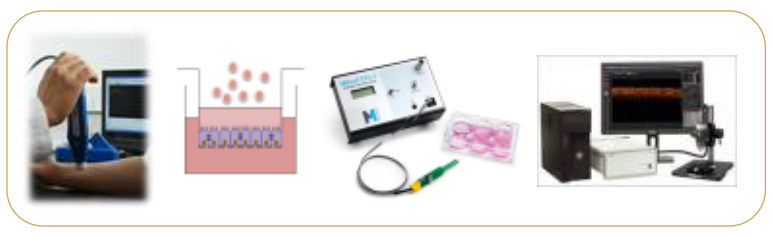

TEER: Reconstructed skin was topically treated with Triton X-100 1%. Tissues were then rinsed with PBS and TEER was measured using EVOM2

Trans-epidermal water loss (TEWL) - was measured directly on the reconstructed skin models using a closed-chamber device, the AQUAFLUX (BIOX)

Permeability coefficient (Kp) on reference chemicals: was quantified by LC/MS/MS to plot the cumulated permeated amount as a function of time

Bodipy®FL permeability: measure with a spectrofluorometer apparatus (Infinite200 Pro, TECAN).

- **OMICS analysis**

Proteins and lipids by high-resolution mass spectrometry

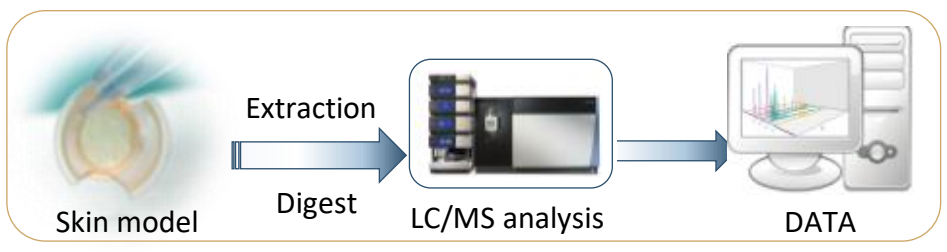
Skin model
Extraction
Digest
LC/MS analysis
DATA

Targeted lipidomics approach focused on free omega fatty acids, free ceramides, bound ceramides extracted from models with different methanol chloroform mixtures was performed by UPLC Vanquish™ coupled to mass spectrometry tribrid IDX™

Untargeted proteomic approach :Tryptic digest analyzed via a Dionex Ultimate™ 3000 nano RSLC coupled to mass spectrometer Orbitrap™ Fusion™.

1. Green H., Kehinde O.,Thomas J. Growth of cultured human epidermal cells into multipleepithelia suitable for grafting. Proc. Natl. Acad. Sci. USA. 1979;76:5665–5668

2. Prunieras M. Epidermal cell cultures as models for living epidermis. J. Investig Dermatol. 1979;73:135–137

3. J van Smeden , M Janssens , G S Gooris , J A Bouwstra The important role of stratum corneum lipids for the cutaneous barrier function . Biochim Biophys Acta. 2014 Mar;1841(3):295-313

4. Schmitt T., Neubert R. State of the art in stratum corneum research. Part II: Hypotehtical stratum corneum lipid matrix models. Skin Pkarmacol Physiol, 2020,33(4):213-230

L'ORÉAL
RESEARCH
& INNOVATION

3 RESULTS & DISCUSSION

- **Barrier function mechanical and physical measurements**

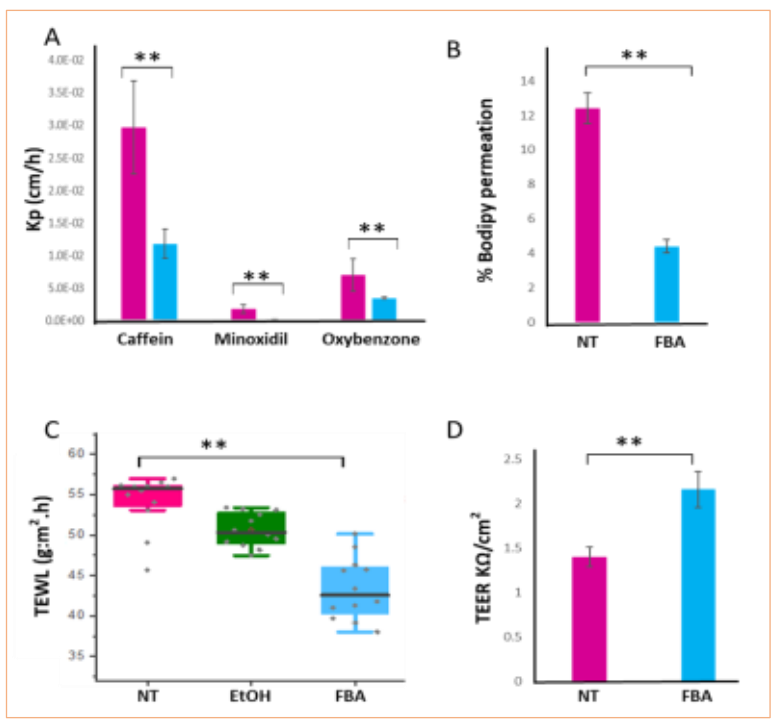
A
Kp (cm/h)
Caffein
Minoxidil
Oxybenzone
B
% Bodipy permeation
NT
FBA
C
TEWL (g:m².h)
NT
EtOH
FBA
D
TEER KΩ/cm²
NT
FBA

Figure 1: reconstructed skin barrier function measurements. A: coefficient of penetration (kp: cm/h) caffein, minoxidil, oxybenzone. B: Bodipy penetration. C: evaluation of transepidermal water loss (TEWL) measurements. D: Transendothelial electrical resistance (TEER) measurement of barrier integrity as an indication of tight junction formation. *=p<0.05, N=3 for each condition

- **OMICS analysis**

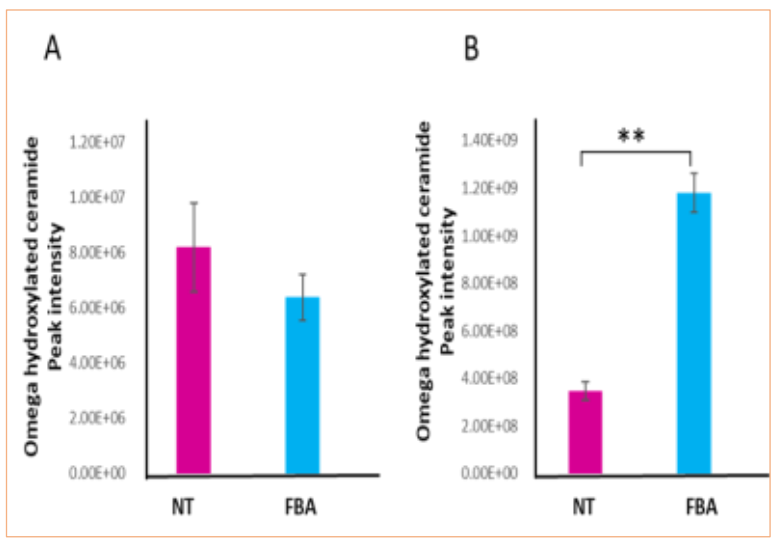
A
Omega hydroxylated ceramide Peak intensity
NT
FBA
B
Omega hydroxylated ceramide Peak intensity
NT
FBA

Figure 2: Free ceramides (A) and protein-bound ceramides (C30 to C34) (B) were measured. Representation of the mean of all 5 omega hydroxylated ceramides is represented (N=6 for each condition) with the standard deviation. **=p <0.05

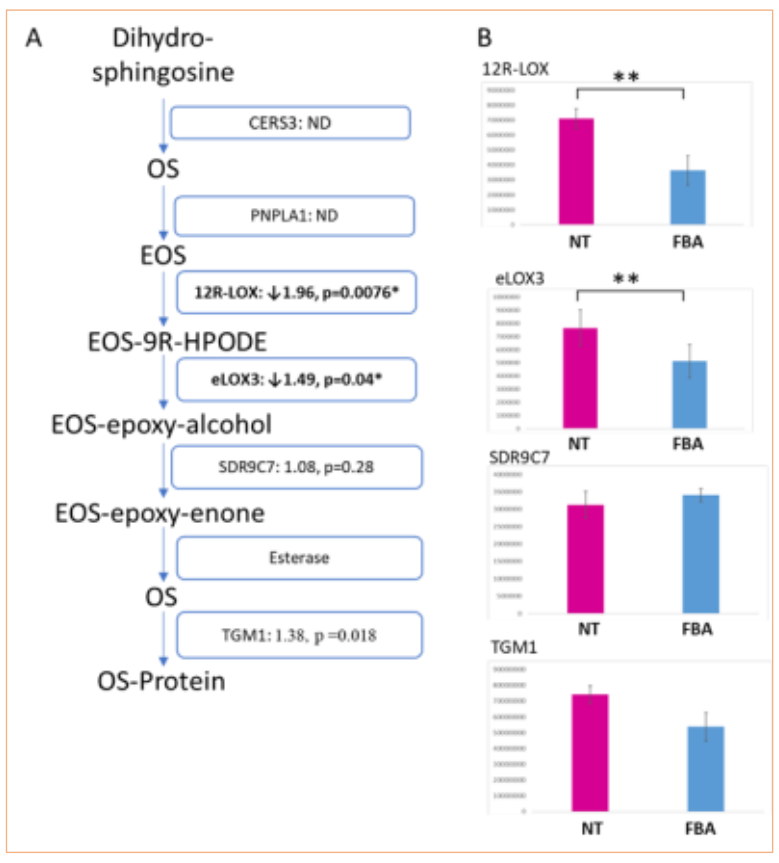
A
Dihydro-sphingosine
CERS3: ND
OS
PNPLA1: ND
EOS
12R-LOX: ↓1.96, p=0.0076*
EOS-9R-HPODE
eLOX3: ↓1.49, p=0.04*
EOS-epoxy-alcohol
SDR9C7: 1.08, p=0.28
EOS-epoxy-enone
Esterase
OS
TGM1: 1.38, p =0.018
OS-Protein
B
12R-LOX
eLOX3
SDR9C7
TGM1
NT
FBA

Figure 3 A) Main metabolic pathway leading to protein-bound ceramide. Enzymes are presented with the fold change, sens of modulation (↓=down regulation) and adj. p. values ND= not detected. B) Histogram representation of the protein abundance based on mean peak intensity (N=5) in the reconstructed skin not treated (NT) and treated with FBA (FBA).

Improved barrier function was observed on the TEWL, TEER and permeation levels demonstrating the positive impact of FBA on SkinEthic™. To decipher the molecular mechanisms responsible for these physical parameters, targeted lipidomics and untargeted proteomics were undertaken. No impact was observed on cholesterol and free ceramides. Significant increased in protein-bound ceramides was associated with the FBA treatment and the physical properties. Focus protein analysis on enzymes leading to protein-bound ceramide revealed significant down expression of 2 important enzyme: 12R-LOX and eLOX3.

4 CONCLUSIONS

Using a multi-parameter analysis, we were able to determine that improved barrier function of reconstructed skins as defined by TEWL, TEER and permeation properties (IN-OUT and OUT-IN) is associated with protein-bound ceramides. The enzymes responsible for the metabolism of protein-bound ceramides are significantly down-regulated at D17.

The increase quantities of protein-bound ceramides is not matched with an increased amount of 12R-LOX and eLOX3 suggesting that either 1- the timing for looking at these enzymes is not optimum and the up-regulation might have happened earlier, which is then counter balance by a down regulation to keep the equilibrium, 2- the activity might be increased while the amount of enzyme is lower, with some compartmentalization involved or 3- other enzymes, not yet identified might participate and in some condition even take over 12R-LOX and eLOX3 activities.

"WHERE BEAUTY, SCIENCE AND INNOVATION MEET!"
32[nd] IFSCC Congress
London 19–22 September 2022

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET