How to deliver to the dermis:
First-in-class dermal delivery technology in human skin.

Bo-Kyung Park1; Won-Seok, Choi1; Young Pil, Jung1; Min-Gyu, Soung1;
1 Cellicon Lab, Rm304, 1662, Yuseong-daero, Yuseong-gu, Daejeon, Republic of Korea

Poster 221

Introduction:

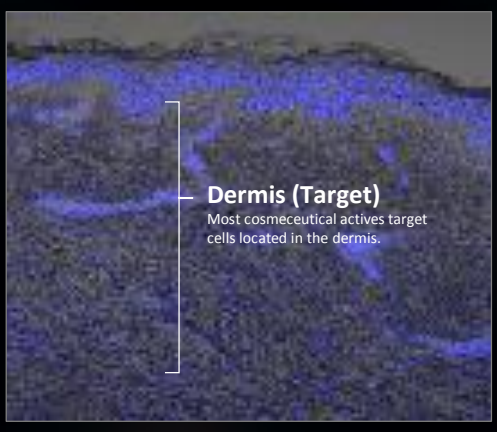
Dermis (Target)
Most cosmeceutical actives target cells located in the dermis.

Problem is that poor penetration shows in cosmeceutical, even it targets the cell in the dermis. Cosmeceutical is expected by many for its outstanding efficacy and mechanism, but its benefits are still controversial. What is certain is that active that target cells in the dermis must reach the dermis.

Challenge is the skin barrier, Stratum Corneum(SC) and Epidermis, that is completely heterogeneous then materials can not reach to the dermis. This limitation is not solved by liposomes or cell-penetrating peptides(CPP). Liposomes can absorb the stratum corneum, but they are not a solution in the epidermal layer composed of living cells. CPPs, on the contrary, are not efficient in the SC.

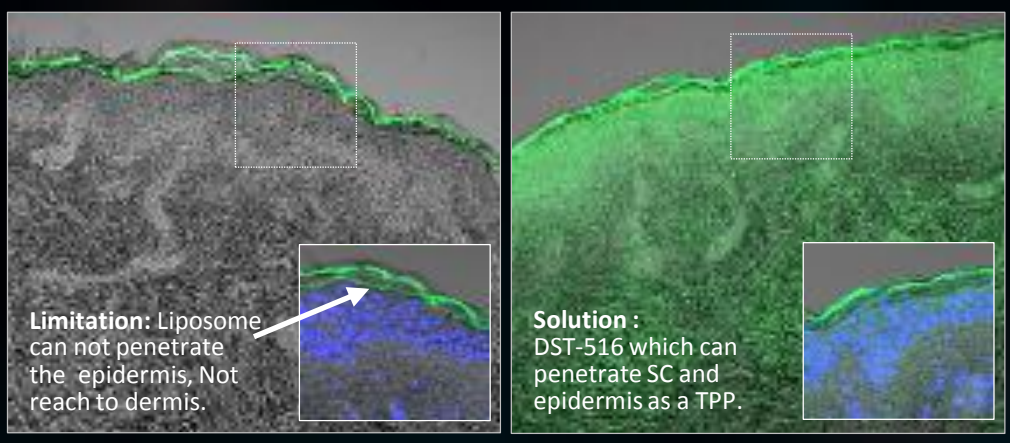
Limitation: Liposome can not penetrate the epidermis, Not reach to dermis.
Solution :
DST-516 which can penetrate SC and epidermis as a TPP.

Solution is a Transdermal Penetrating Peptide(TPP) which has a both liposome and CPP properties. This study reports a novel peptide which has capability to deliver to dermis without covalent bond with various cargo and it is demonstrated by confocal microscope. Furthermore, we aimed to demonstrate the pathway of skin permeation in the SC and the epidermis.

Conclusions:

1. DST-516 (Oligopeptide-191) as TDS Platform DST-516 delivered various cargoes with different sizes and physicochemical properties to the dermis in this study.

2. PK analysis on delivery of DST-516 A Pharmacokinetics study of dermal transfer by DST-516 was conducted. It was confirmed that DST-516 passed through the intercellular pathway in the SC of the skin and the intracellular pathway in the epidermal layer to reach the dermis and showed rapid permeation between 1.5 hour to 2 hours.

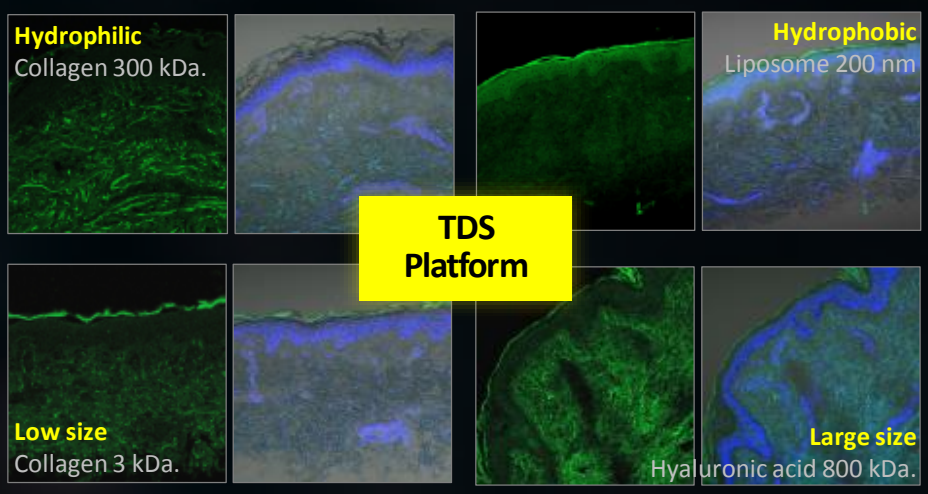
Hydrophilic
Collagen 300 kDa.
Hydrophobic
Liposome 200 nm
TDS Platform
Low size
Collagen 3 kDa.
Large size
Hyaluronic acid 800 kDa.

Materials & Methods:

Oligopeptide-191 were synthesized using solid-phase peptide chemistry. Skin penetration are evaluated using cadaver skin of full thickness and using confocal scanning laser microscopy. Section images are observed 3 hours after cargo treated.

Results & Discussion:

1. Delivery efficacy of various cargoes

In order to examine the possibility of platform TDS of DST-516, the dermal transfer effect was tested as a cargo of various Cosmetic raw materials. It was confirmed that DST-516 can deliver dermis from low molecules to polymers.

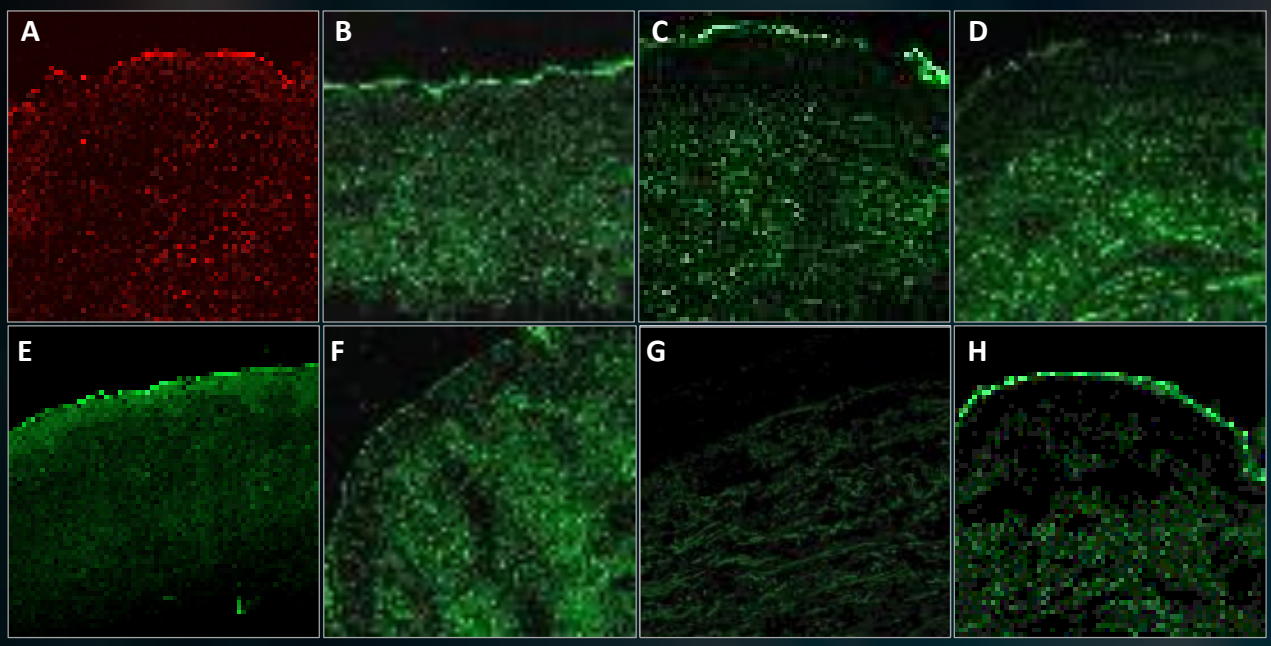
A
B
C
D
E
F
G
H

Figure. Confocal image of DST-516 (Texas red) and various cargos assembled with DST-516. Image captured washed-off 3 hours after treatment A: DST-516 only, B-D: Collagen (3k, 30k, 300 kDa), E: 200 nm liposome, F: Hyaluronic acid (800 kDa), G: EGF, H: Tranexamic acid.

2. Pathway on skin permeation

SC : Intercellular lipid / Epidermis : Intracellular transmission (Cell to cell)

To investigate transdermal pathway, confocal was conducted on peeling-off cadaver skin and keratinocyte.

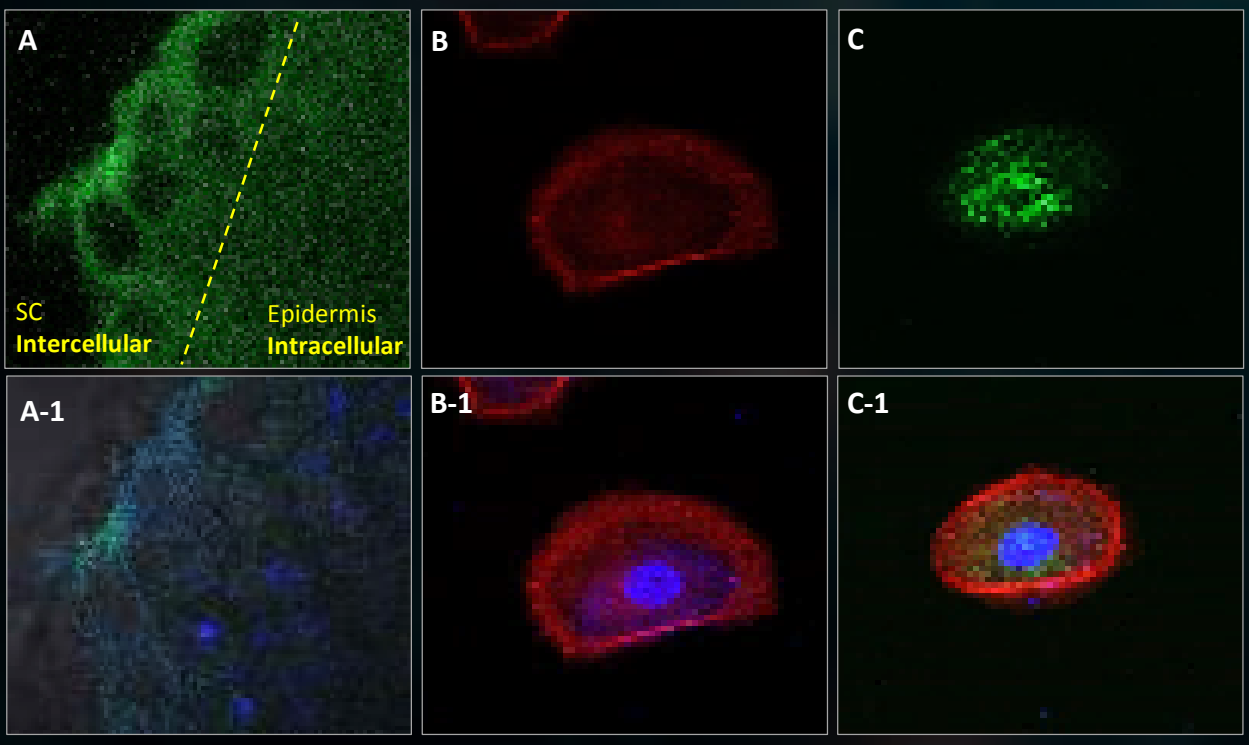
A
SC
Intercellular
Epidermis
Intracellular
B
C
A-1
B-1
C-1

Figure. Pathway on skin permeation (A) and Verification of epidermal pathway by keratinocyte permeability. (B and C). Figure A. Treated FITC-hyaluronic acid onto skin then SC and epidermis were peeled off the skin and frozen section slides are prepared. (A: Merged, B: Confocal) Figure B and C. Texas red DST-516 and FITC-hyaluronic are treated to HaCaT Cell. (B: DST-516 only, C: FITC-hyaluronic acid and DST-516 co-treated).

3. PK analysis of delivery

Delivery rate of DST-516 and the hyaluronic acid with DST-516 was quantitatively analyzed using confocal intensity by time. As a result of it the dermal layer with fluorescent intensity rapidly increased after 1 hour half, and this pattern was observed in the DST-516 only group and with HA.

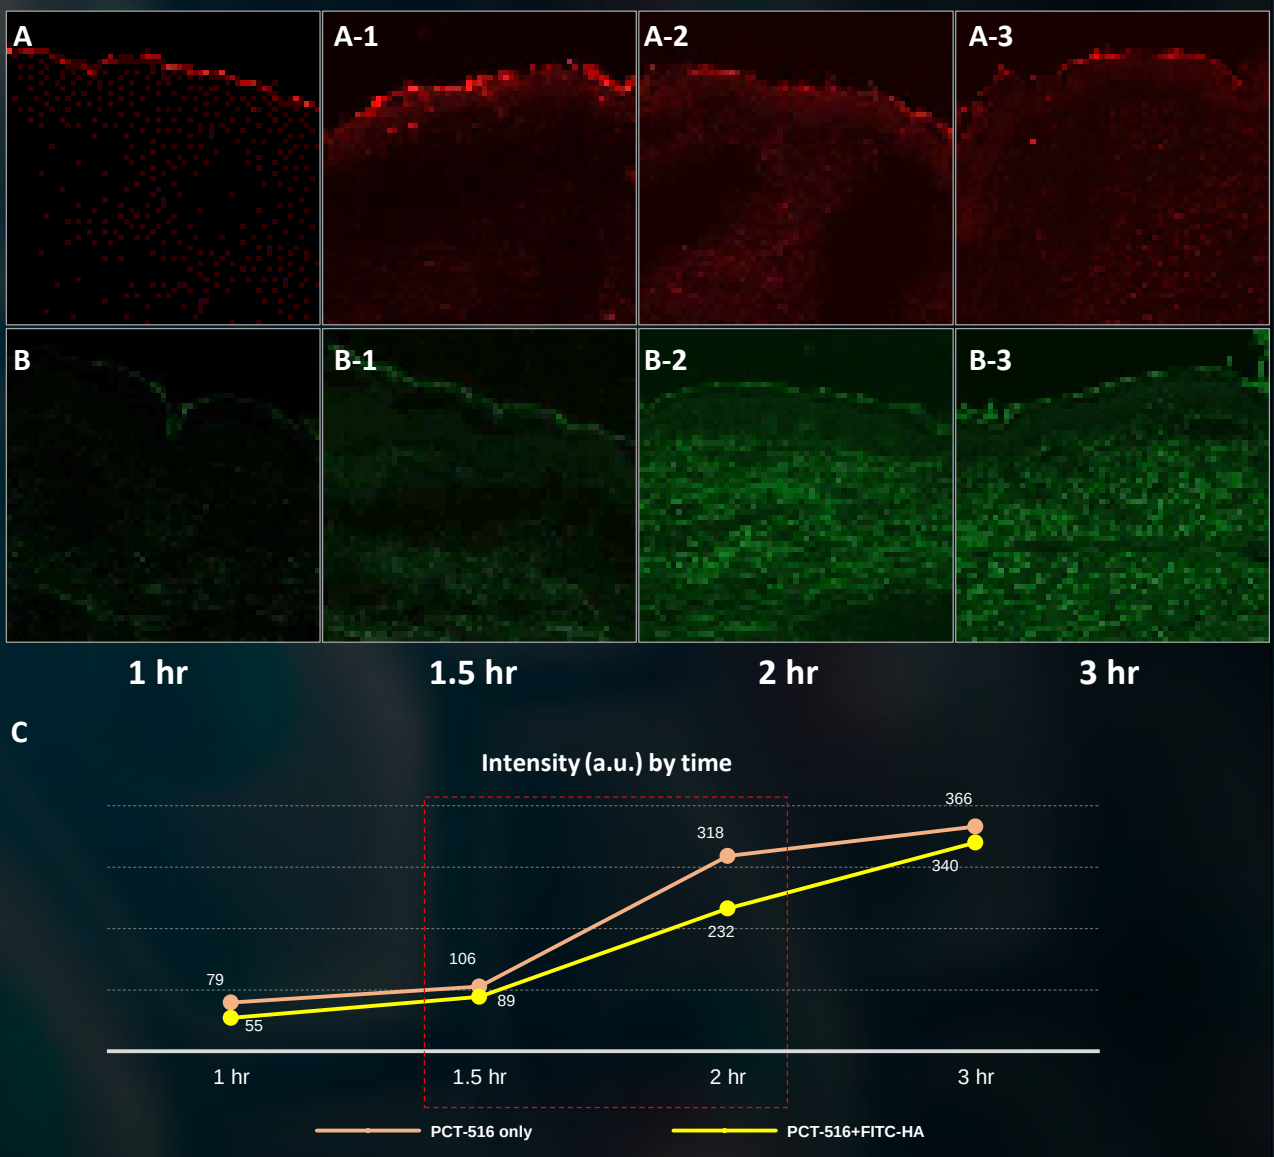
A
A-1
A-2
A-3
B
B-1
B-2
B-3
1 hr
1.5 hr
2 hr
3 hr
C
Intensity (a.u.) by time
79
55
106
89
318
232
366
340
1 hr
1.5 hr
2 hr
3 hr
PCT-516 only
PCT-516+FITC-HA

Figure. Dermal delivery rates of DST-516 and DST-516 + HA by time, A. DST-516 texas, B. FITC-HA with DST-516, C. Intensity (a.u.) by time

4. Synergy for promoting collagen systhesis in fibroblast of DST-516 by cell penetrating efficacy

Collagen production promotion of cargo (Peptide, HA) combined with DST-516 was tested. Two cargoes that stimulate fibroblast to promote collagen production with DST-516 was significantly increased.

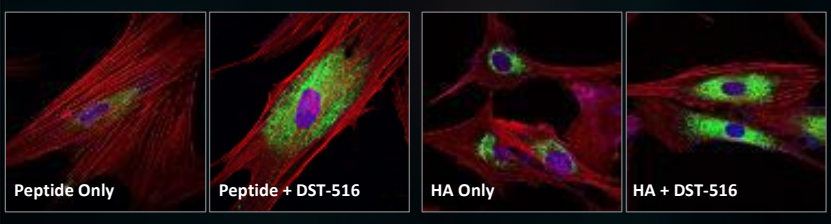
Peptide Only
Peptide + DST-516
HA Only
HA + DST-516

Red: HDF actin, Green: type I collagen anti-body.

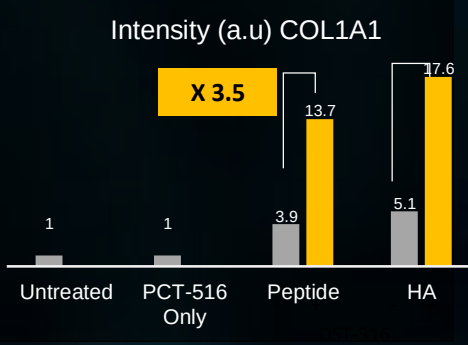
Intensity (a.u) COL1A1
X 3.5
1
1
3.9
13.7
5.1
17.6
Untreated
PCT-516 Only
Peptide
HA

References:

1. Ji-Yeon Chung, Hyo-Sun Han, (2014) The Recent Trend of Percutaneous Absorption Used in Cosmetics. Kor. J. Aesthet. Cosmetol 12(5):597-605.
2. M. R. Prausnitz and R. Langer, (2008) Transdermal drug delivery. Nat. Biotech., 26(11):1261-1268.
3. A. Eguchi, T Akuta, et al (2001) Protein transduction domain of HIV-1 Tat protein promotes efficient delivery of DNA into mammalian cells, J. Biol. Chem., 276(28), 26204-26210.
4. Howe CL (2005). Modeling the signaling endosome hypothesis: why a drive to the nucleus is better than a (random) walk. Theoretical Biology & Medical Modelling 2 (1):43.
5. Aung Than,Ke Liang, et al (2017) Transdermal Delivery of Anti-Obesity Compounds to Subcutaneous Adipose Tissue with Polymeric Microneedle Patches. Small Methods 1(11): 1700269.

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET