SINCERE AND RELIABLE SPF: A QUALITY PROCESS THAT CAN BE DEPLOYED WORLDWIDE

#449

Piquemal, Pascale[1]; Renoux, Pascale[1]; Clemente, Dáphine Antônia[2]; Salgado-Santos, Idalina[3]; Pouradier, Florence[4]

L'Oréal Research & Innovation, Evaluation Intelligence Dpt; [1]Chevilly-Larue, France; [2]Rio de Janeiro, Brazil, [3]Clark, USA; [4]Clichy,France

1 INTRODUCTION

Sunlight can be harmful to all skin tones when adequate protection is not used. Many biological and clinical effects are associated with the improper sun exposure and the use of sunscreen may prevent the skin damage. The level of sun protection provided by a product has traditionally been proven using standardized and harmonized in vivo methods **[1;2]**. These standards provide a controlling frame regarding the different aspects of the method: required equipment specifications, test subject inclusion criteria, use of reference samples, sunscreen application, UV exposure, erythema readings and procedure to determine the SPF. However, despite the standardization, variability may be observed in the results **[3]**. This variability could be explained by different experimental conditions, factors in volunteers' selection, product application procedures, visual assessment of erythema. In this context, the quality process is a mandatory part, preparing for all critical steps aimed at improving the quality, safety and efficacy of sunscreens as an important part of consumer protection against UV damages.

The intention of this communication is to describe the quality process our Group developed alongside our partners. It is a highly collaborative work effort we are using to achieve the sincerest and most reliable SPF globally, with a high level of quality, and respecting the different local regulations.

2 MATERIALS & METHOD

The photoprotection quality process is based on two important strategies illustrated below:

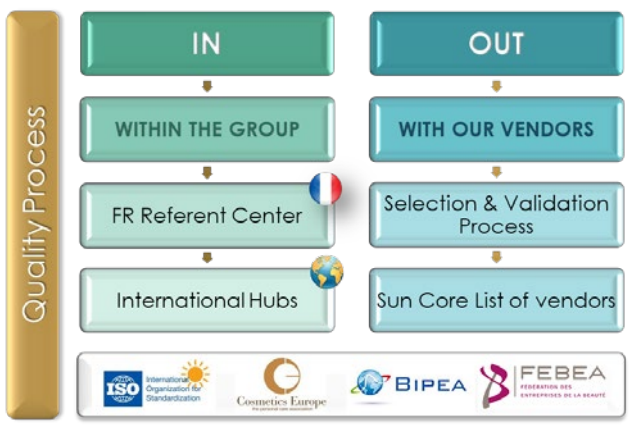
Quality Process
IN
OUT
WITHIN THE GROUP
WITH OUR VENDORS
FR Referent Center
Selection & Validation Process
International Hubs
Sun Core List of vendors
ISO
Cosmetics Europe
BIPEA
FEBEA

1/ Internally (IN): an international Photoprotection Community created within the R&I teams of our Group with strong local anchorage supported by a referent center created more than 30 years ago and whose expertise is recognized worldwide. This community built and deployed an international vision and a global strategy of testing, respecting the local regulations and following a common objective of quality and sincerity of claims for consumer protection. Key points were to build a global expertise and an international alignment in evaluation strategy: common process of selection, validation of a new and maintenance of the quality level of the validated vendors over the time.

2/ Externally (OUT): working with the CROs, considering each key points of the SPF determination process, in line with both ISO and FDA standards:

Key steps	Validation and surveillance points
Inclusion of volunteers	skin color homogeneity, test area quality
Dispensing and formula application	checking the spreading and homogeneity of the product and having technicians trained and validated on the proper application (first "digital" monitoring, final validation in situ)
UV Exposures and controls	measurements of fluxes before each exposure,
Readings	Initial train and validation using MED atlases, then final validation in situ for each technician. Maintain with ring tests.
System quality audit	quality process checking including compliance obligations and adjustments in terms of Good Practices, with emphasis on GLP and GCPs
Final Validation Process confirmation	blinded study using internal reference formulas, observing all steps described, as well as the final SPF results expected for our internal reference.
Surveillance	following an internal action plan, formulas whose SPF are well known are regularly added, in order to validate the test results for the products **[4]**
Quality Maintenance	periodic training, monitoring, audits and ring tests are kept used as part of Photoprotection CROs Quality Plan.

3 RESULTS & DISCUSSION

Our community identified and deployed a robust quality process which covers each of the method specifications required in both ISO and FDA standards. Many checkpoints are incorporating in, taking the best of each method as long as it was consistent within the other. For example, for the inclusion of test subjects, we consider the homogeneity of skin color and the quality of the test area using ITA° as requested in ISO24444, which is not specified by the FDA method today but is completely within its framework, nevertheless.

To deploy this process in a new CRO for integrating them into the Core List requires time and resources from both parts. The validation is done by method and by technician. On average we devote over one year for a new CRO to complete the full process. Most of the steps were developed to be compatible with remote monitoring conditions, yet the final validation is still done in situ.

The impact of this process on reducing the results variability is unfortunately quite difficult to evidence. Before this deployment we missed key quality indicators, other than the use of the sunscreen reference product requested in the standards, which is an important yet limited way to validate a test result. The absence of such indicators is a strong limitation for quantifying the impact of our process, and we are working toward defining some.

This procedure induces positive influence for all parties, generating a virtous circle as illustrated below:

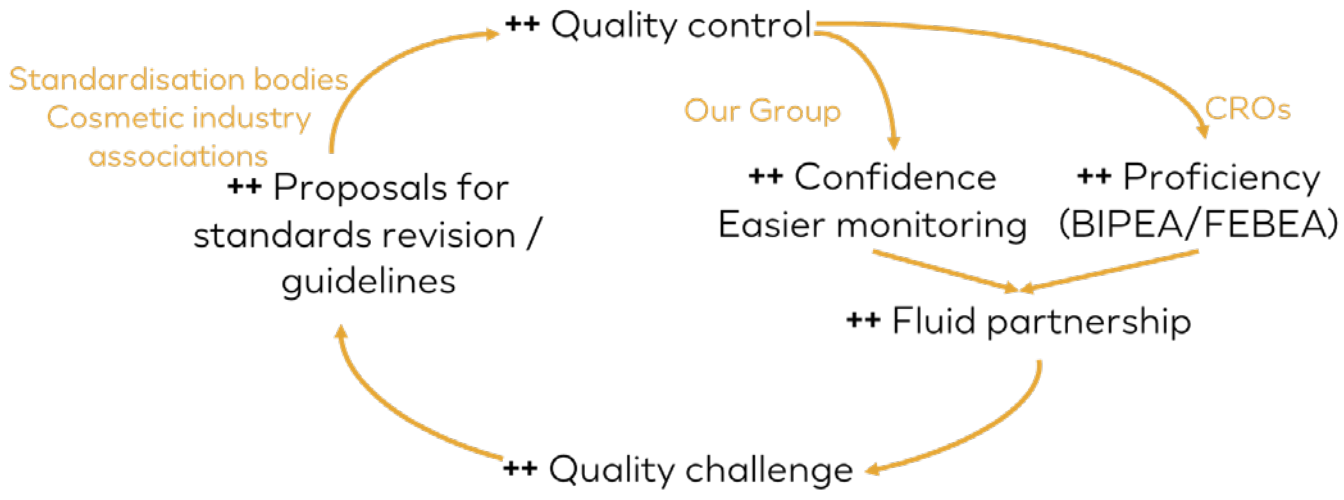
++ Quality control
Standardisation bodies
Cosmetic industry
associations
Our Group
CROs
++ Proposals for standards revision / guidelines
++ Confidence
Easier monitoring
++ Proficiency
(BIPEA/FEBEA)
++ Fluid partnership
++ Quality challenge

To formalize this approach and share these results with all the international teams in charge of conducting SPF testing, a "Sun Barometer" of CROs was deployed, and regularly updated thanks to systematic monitoring done of the CROs of the Core List. This document aims to gather and summarize all information relevant to our product being tested in reliable conditions, including but not limited to the regulatory frame for each country, which CRO is qualified to perform which method, and where they are in the validation and/or maintenance processes.

4 CONCLUSIONS

This quality guidance put in place by our Group, in partnership with our vendors, allows us to enhance the reliability of the SPF worldwide. We accomplish this while respecting local regulation and deploying a high level of quality control, thus avoiding any risk to the consumer's health, and results in claiming sincere SPF. This quality system, which we described here for the SPF measurement, is applicable for the other in-vivo photoprotection indices and methods, such as UVA-PF and Water-resistance. Additionally, it is a useful source for analyzing the functioning and improvements to the process, providing a more accurate identification of risks, and leading to the development of more effective action plans in the future.

REFERENCES

1. ISO 24444: 2010 Cosmetics–Sun Protection Test Methods – In vivo Determination of the Sun Protection Factor (SPF)

2. FDA 2011 Labeling and Effectiveness Testing; Sunscreen Drug Products for Over the Counter

3. Miksa, S., et al. (2016). "Sunscreen sun protection factor claim based on in vivo interlaboratory variability." Int J Cosmet Sci 38(6): 541-549.

4. Renoux, P. et al (2022). « How to reinforce the proficiency of SPF testing". IFSCC London 2022.

L'ORÉAL RESEARCH & INNOVATION

ACKNOWLEDGMENTS

The authors express their acknowledgments to all the internal Photoprotection Community members worldwide and to the whole CROs network for their commitment in the present quality process.

"WHERE BEAUTY, SCIENCE AND INNOVATION MEET!"
32nd IFSCC Congress
London 19–22 September 2022

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET