HOW TO REINFORCE THE PROFICIENCY OF SPF TESTING

#439

Renoux, Pascale[1]; Clemente, Dáphine Antônia[2]; Piquemal, Pascale[1]; Pouradier, Florence[3]

L'Oréal Research & Innovation, [1] Chevilly-Larue, France; [2]Rio de Janeiro, Brazil, [3]Clichy, France

1 INTRODUCTION

As photoprotection is about public health, sunscreen labelling must be sincere and reliable. Performance on UVB or UVA protection (expressed through SPF or UVA-PF) and Water-resistance is determined through standardized methods. Considering the importance of controlling the quality of the performed determination, all these standards must use sunscreen formulation reference to validate the test, on each of the volunteers included in the clinical test **[1]**. As an interesting initiative, one of these reference sunscreens (P2) was used through meta-analysis to leverage knowledge on SPF methods comparison or impact of the time of year, gender, age or Fitzpatrick's skin type of the volunteers on the measured SPF **[2]**.

To reinforce this quality control approach, we chose to also include regularly in our photoprotection studies well-known formulas, representative of marketed products.

Through this approach, we can collect a large amount of data, from different laboratories, worldwide. The objective of this study was to investigate how these data can be exploited, beyond the one-point approach test by test, to reinforce our global quality process and leverage knowledge on testing of the same formula across different world populations.

2 MATERIALS & METHODS

To ensure the reliability of the sun protection values of our main projects, and as part of our CROs quality control process **[3]**, we decided to include reference formulas representative of our catalogue in some in vivo SPF studies, besides the standards formulas recommended by the specific norm. The 3 chosen formulas are representative from the market and present different level of protection.

Labelled category	Data	Number of CROs	Countries
Medium Protection	227 individual data from 42 studies	7	Europe (4 countries) Canada Singapore
High protection	1279 individual data from 231 studies	15	Europe (7 countries) Canada Brazil China Japan Singapore
Very high protection	1931 individual data from 360 studies	12	Europe (7 countries) Canada Brazil China Japan Singapore

Table 1. Description of the database on the 3 reference formulas tested worldwide. Some European countries don't have enough data to be relevantly included in the analysis. China, Japan and Singapore are artificially merged to build an "Asia" area.

Statistical analysis of this database is performed for each study and time by time, both to identify a criterion which reinforces the SPF determination and to evaluate the impact of the studied population on the obtained results.

For success criteria, inspired by BIPEA's approach and referring to the ISO 13528 standard **[4]**, we used the "z-score" which is continuously updated by considering the global data from a formula, to calculate an accepted range.

The z-score (z) is calculated from the laboratory result (x) based on the assigned value (x_{pt}) and half the tolerance value :

$$z = (x - x_{pt}) / (VT/2).$$

ISO13528 standard defines that its absolute value greater than 2.0 is equivalent to a warning signal, while an absolute value greater than or equal to 3.0 is considered an action signal.

3 RESULTS & DISCUSSION

The z-score revealed to be a relevant and useful criterion to assist the study sponsor in validating the reliability of the obtained results or alert on the need to put in place an action plan with the CRO or on the necessity of a new training.

When a study presents a z-score out of the acceptable limit, an investigation and exchange with the CRO in charge of this study is started to understand which parameter may have caused the unexpected result such as:
- how much equipment was used and whether it was calibrated.
- who were the technicians who conducted the study and whether they are validated within the framework of our quality process **[3]**.
- if an unexpected event occurred during the study

If after such investigation, the CRO continues to present continuously too high absolute value for z-scores, we dismissed it temporarily. Then, new application and reading trials are started for each of the technicians and a new ring study is performed. If the results are acceptable, the CRO is reinstated. Such event can be visualized in Figure 1:

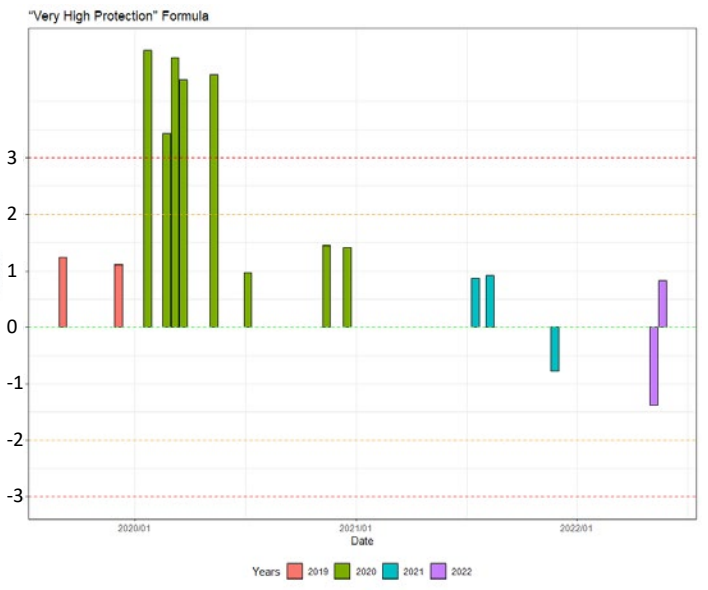

"Very High Protection" Formula
z-score
3
2
1
0
-1
-2
-3
2020/01
2021/01
2022/01
Date
Years 2019 2020 2021 2022

With our database, even if some geographical areas are represented by one or two laboratory(ies) only, the repetition of the testing and the quantity of the data collected allow us to investigate the population effect.

To avoid the results being biased by under- or over-estimated values, only the studies with a |z-score| < 2.0 were included in the analysis. Data are visualized in Boxplots and the geographical area effect is investigated by ANOVA and T-tests to estimate the p-value, plus Bayesian approach to estimate the effect-size.

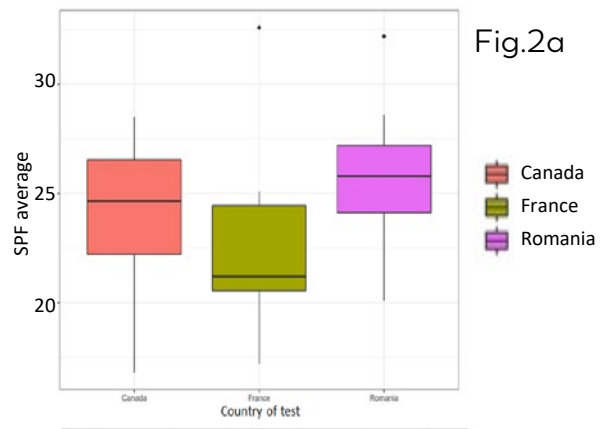

SPF average
30
25
20
Country of test
Canada
France
Romania

Fig.2a

For the "medium Protection" formula, no statistical difference is observed between Canada (North America), France and Romania (Europe) (Fig.2a).

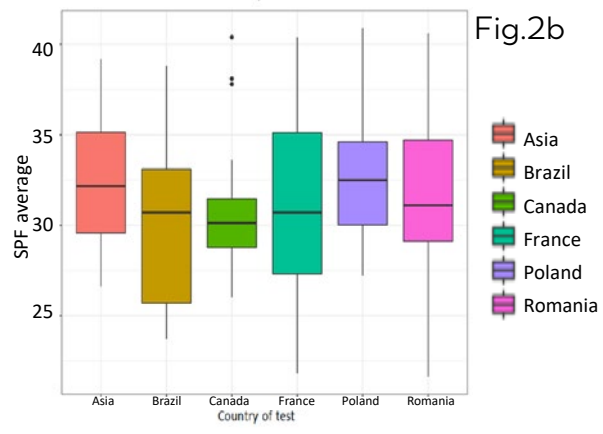

SPF average
40
35
30
25
Asia
Brazil
Canada
France
Poland
Romania
Country of test

Fig.2b

For the "high Protection" formula, no statistical difference is observed between Asia, Brazil (South America), Canada (North America), France, Poland and Romania (Europe) (Fig.2b).

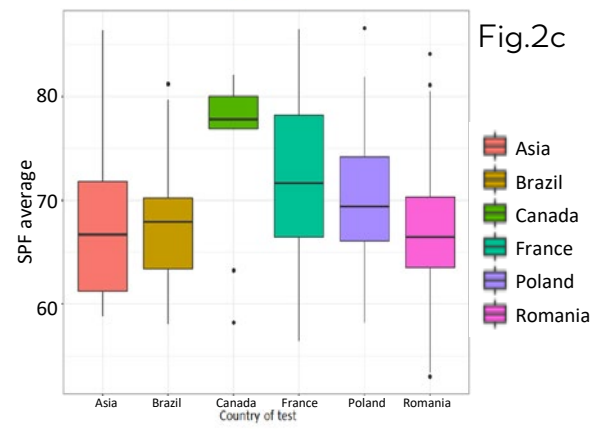

SPF average
80
70
60
Asia
Brazil
Canada
France
Poland
Romania
Country of test

Fig.2c

For the "very high protection" formula we observed a higher SPF value in Canada (North America) than in Asia, Brazil (South America) and Romania (Europe) with a moderate effect-size (Fig.2c). We hypothesize this difference not being clinically relevant, but due to unbalanced sampling across the countries. Our database should be reinforced to be able to confirm or reject this hypothesis

4 CONCLUSION

The sun protection field is continuously moving for higher and higher reliability. However, it does not replace internal quality control or an assessment of compliance with standards. Moreover, data quality assessment is an important part of the overall quality management system.

The approach described in this poster, allows us to control both the instantaneous performance and the continuous performance of the laboratory, ensuring the robustness of the SPF determination. The analysis of the resulting database highlights the consistency of the value obtained from different population across the world on which this standardized determination is performed.

REFERENCES

1. *ISO 24444:2019 Cosmetics–Sun Protection Test Methods – In vivo Determination of the Sun Protection Factor (SPF)*

2. *Alejandria, M., et al. (2019). "Disparate SPF Testing Methodologies Generate Similar SPFs. II. Analysis of P2 Standard Control SPF Data. » Journal of cosmetic science 70(4): 181-196.*

3. *Piquemal, P., et al. (2022). "Sincere and reliable SPF: a quality process that can be deployed worldwide." Poster. IFSCC 2022 London.*

4. *ISO13528:2015 Statistical methods for use in proficiency testing by interlaboratory comparison*

L'ORÉAL RESEARCH & INNOVATION

ACKNOWLEDGMENTS

The authors express their sincere gratitude to Hicham Nocairi for the statistical support, and Remi Moyenin for the support in data management, as well as all the study managers in the L'Oréal R&I hubs worldwide who help collecting these data.

'WHERE BEAUTY, SCIENCE AND INNOVATION MEET!' 32nd IFSCC Congress London 19–22 September 2022

32ND IFSCC CONGRESS, LONDON 2022 - WHERE BEAUTY, SCIENCE AND INNOVATION MEET